Expanding the Serotype Frontier: Design of Synthetic AAV Capsids with Artificial Intelligence

Sam Sinai, David Brookes, Jeffrey Chan, Alex Mijalis, Alex Kern, Neha Prasad, Theodore Sternlieb, Farhan Damani, Meghan Lambie, Viktoria Betin, The Dyno Therapeutics Team, Kathy Lin, Jamie Kwasnieski, Adrian Veres, Eric Kelsic; Dyno Therapeutics, Watertown, MA, USA 02472

Overview

- Many patients are ineligible for gene therapies due to pre-existing immune recognition of naturally occurring Adeno-associated virus (AAV) capsids.
- The number of known natural AAV serotypes is limited, slowing the discovery of clinically useful new serotypes.
- Advances in generative AI have enabled the design of synthetic AAV capsids that are significantly different from existing serotypes, opening a new route for the discovery of therapeutically relevant AAVs.
- Here we show that AI models can generate functional capsids with many novel mutations that are unlikely to arise through natural or directed evolution.
- We validate designs of up to 50 mutations at once, introducing changes in distinct structural components of AAV9.
- A number of these diverse capsids show enhanced in vitro transduction and immune evasion relative to AAV9 suggesting a promising future direction for widespread availability of gene therapy and potential for re-dosing.

Synthetic AAVs are diverse in sequence and structure

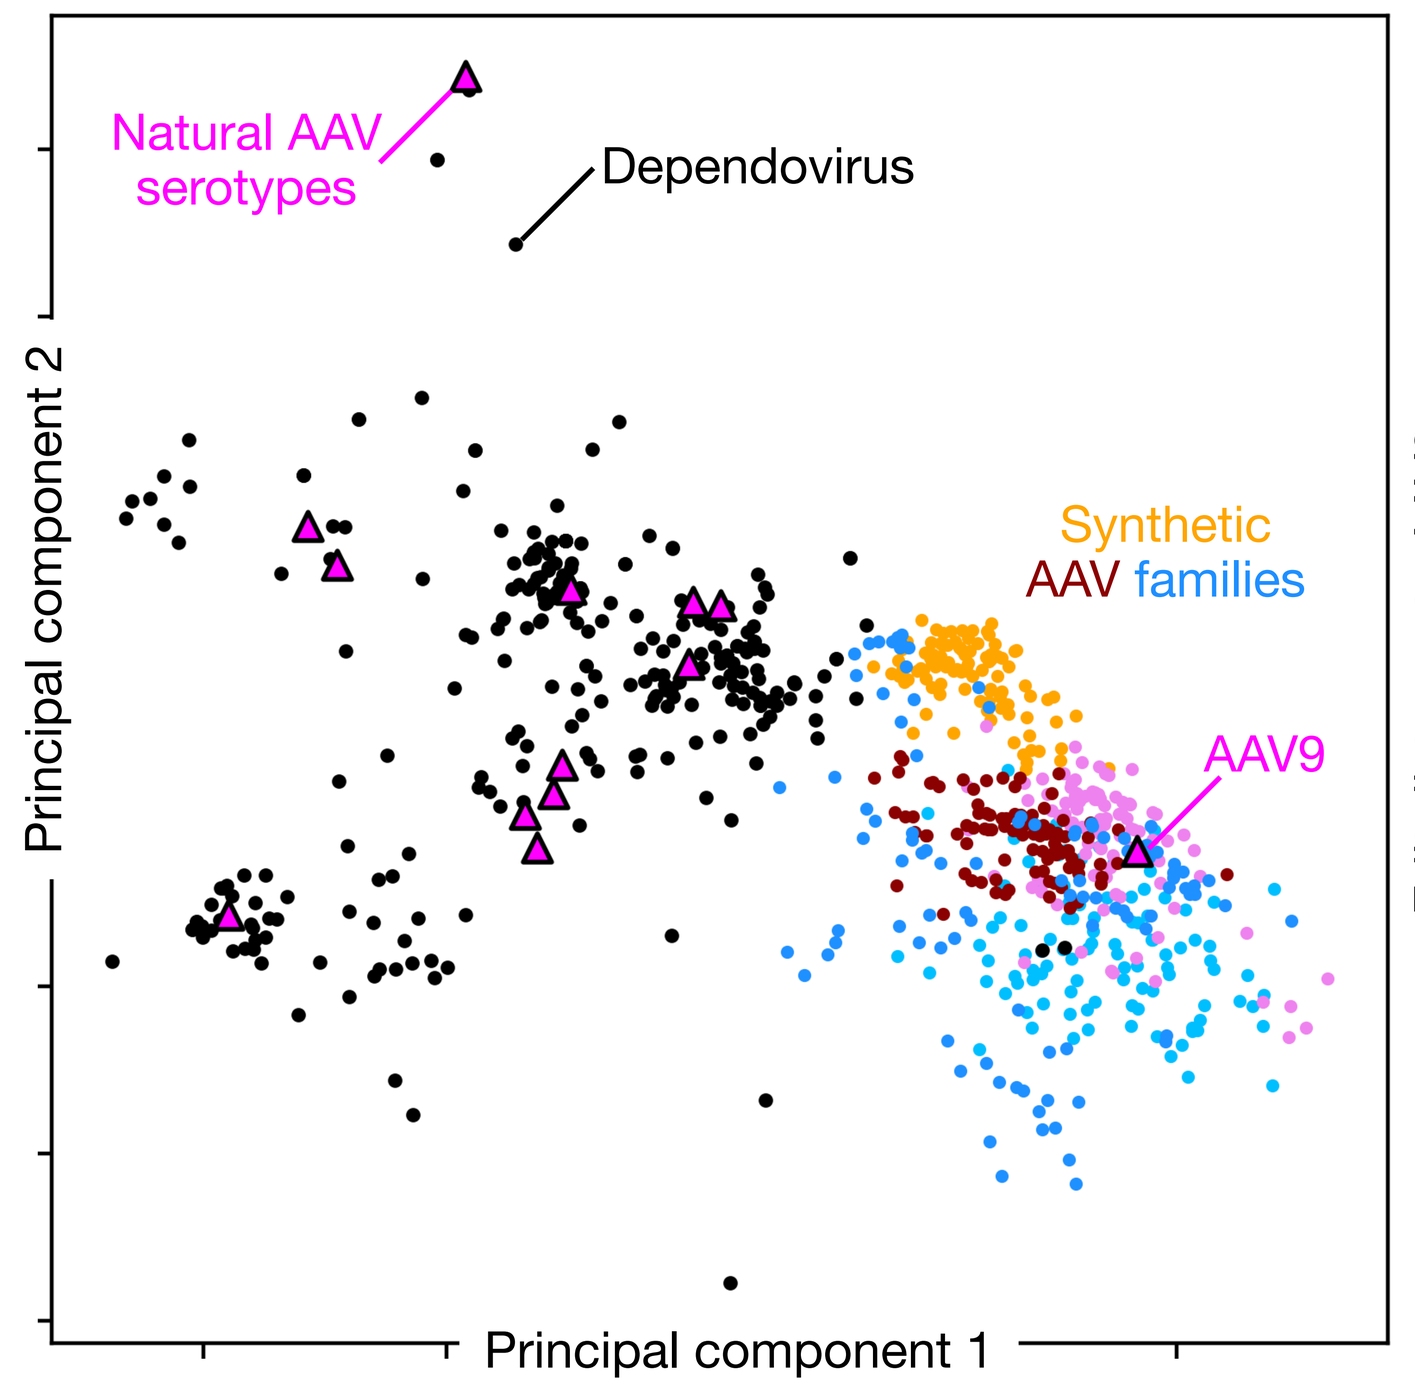
Natural AAV serotypes
Dependovirus
Synthetic AAV families
AAV9
Principal component 2
Principal component 1

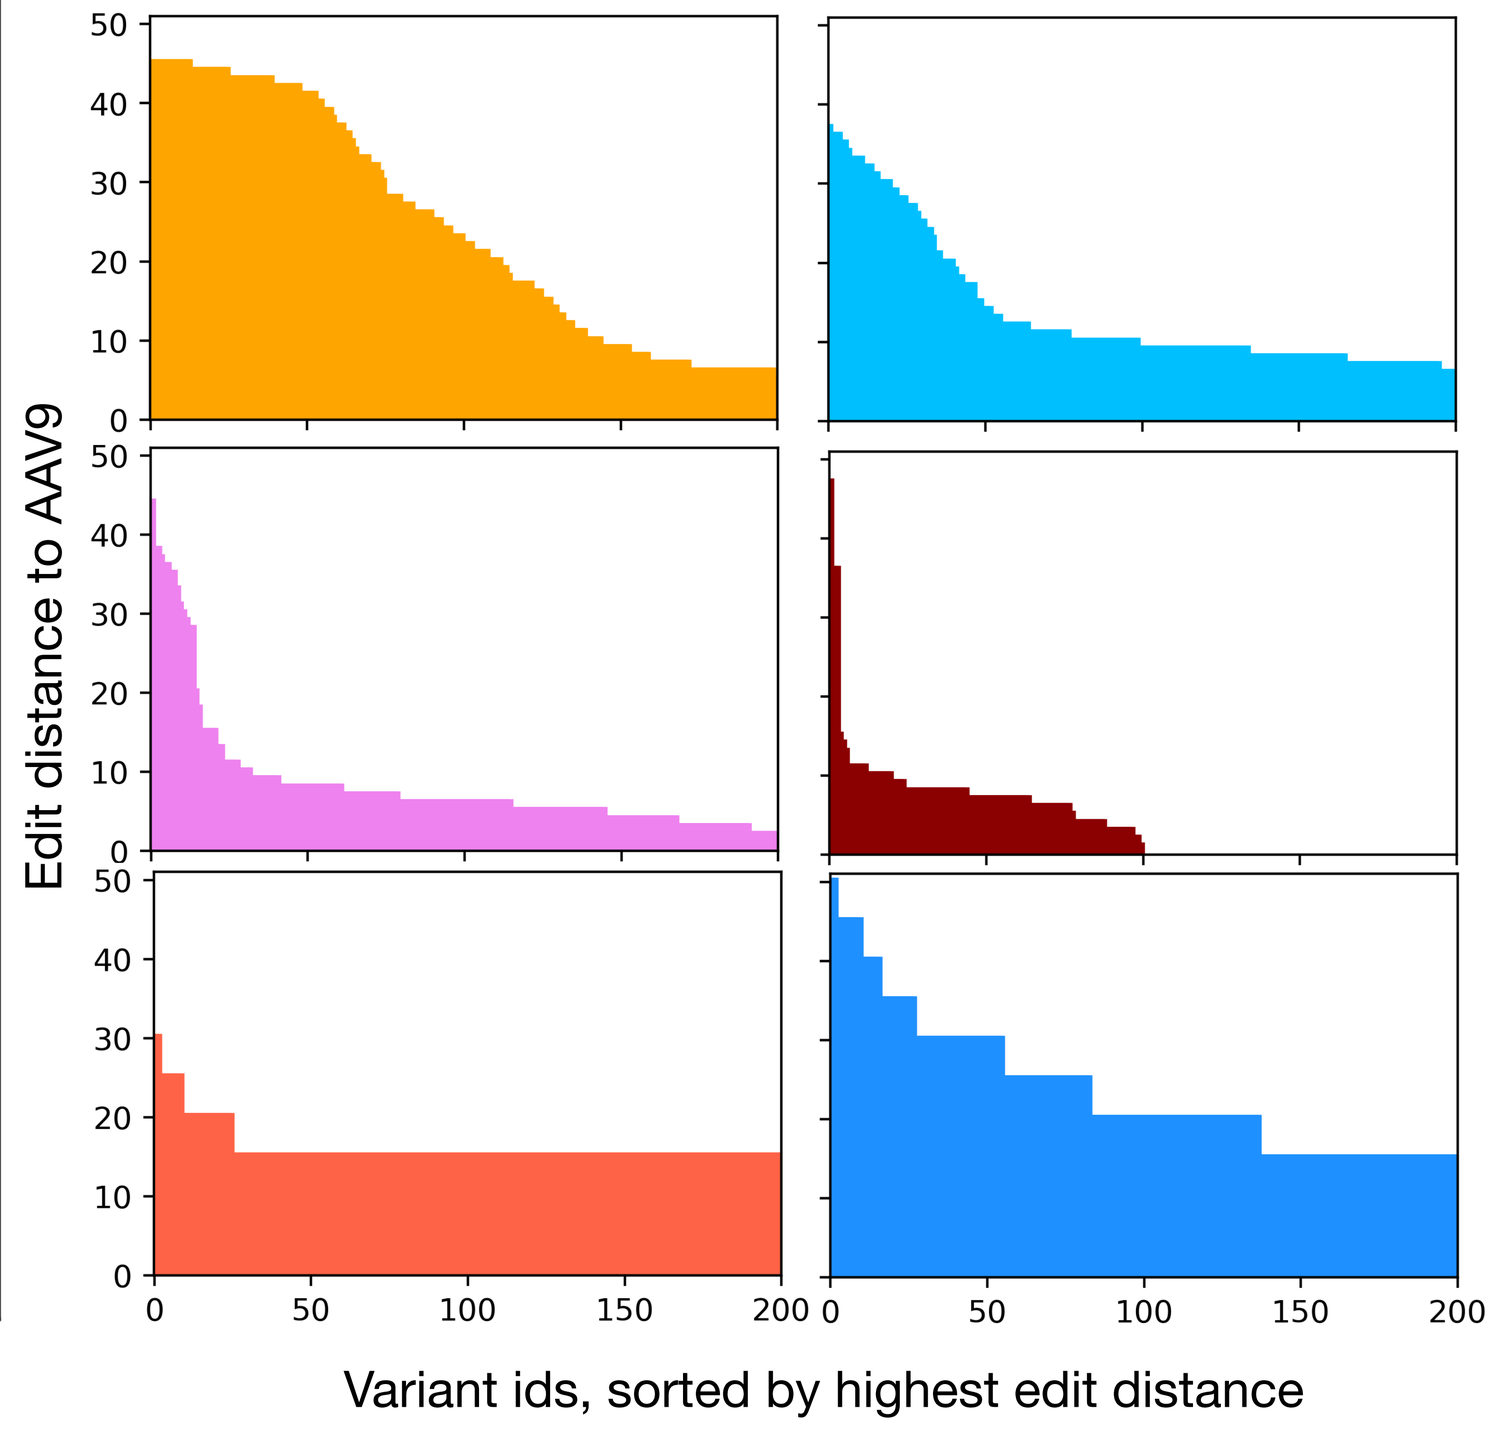
Edit distance to AAV9
Variant ids, sorted by highest edit distance

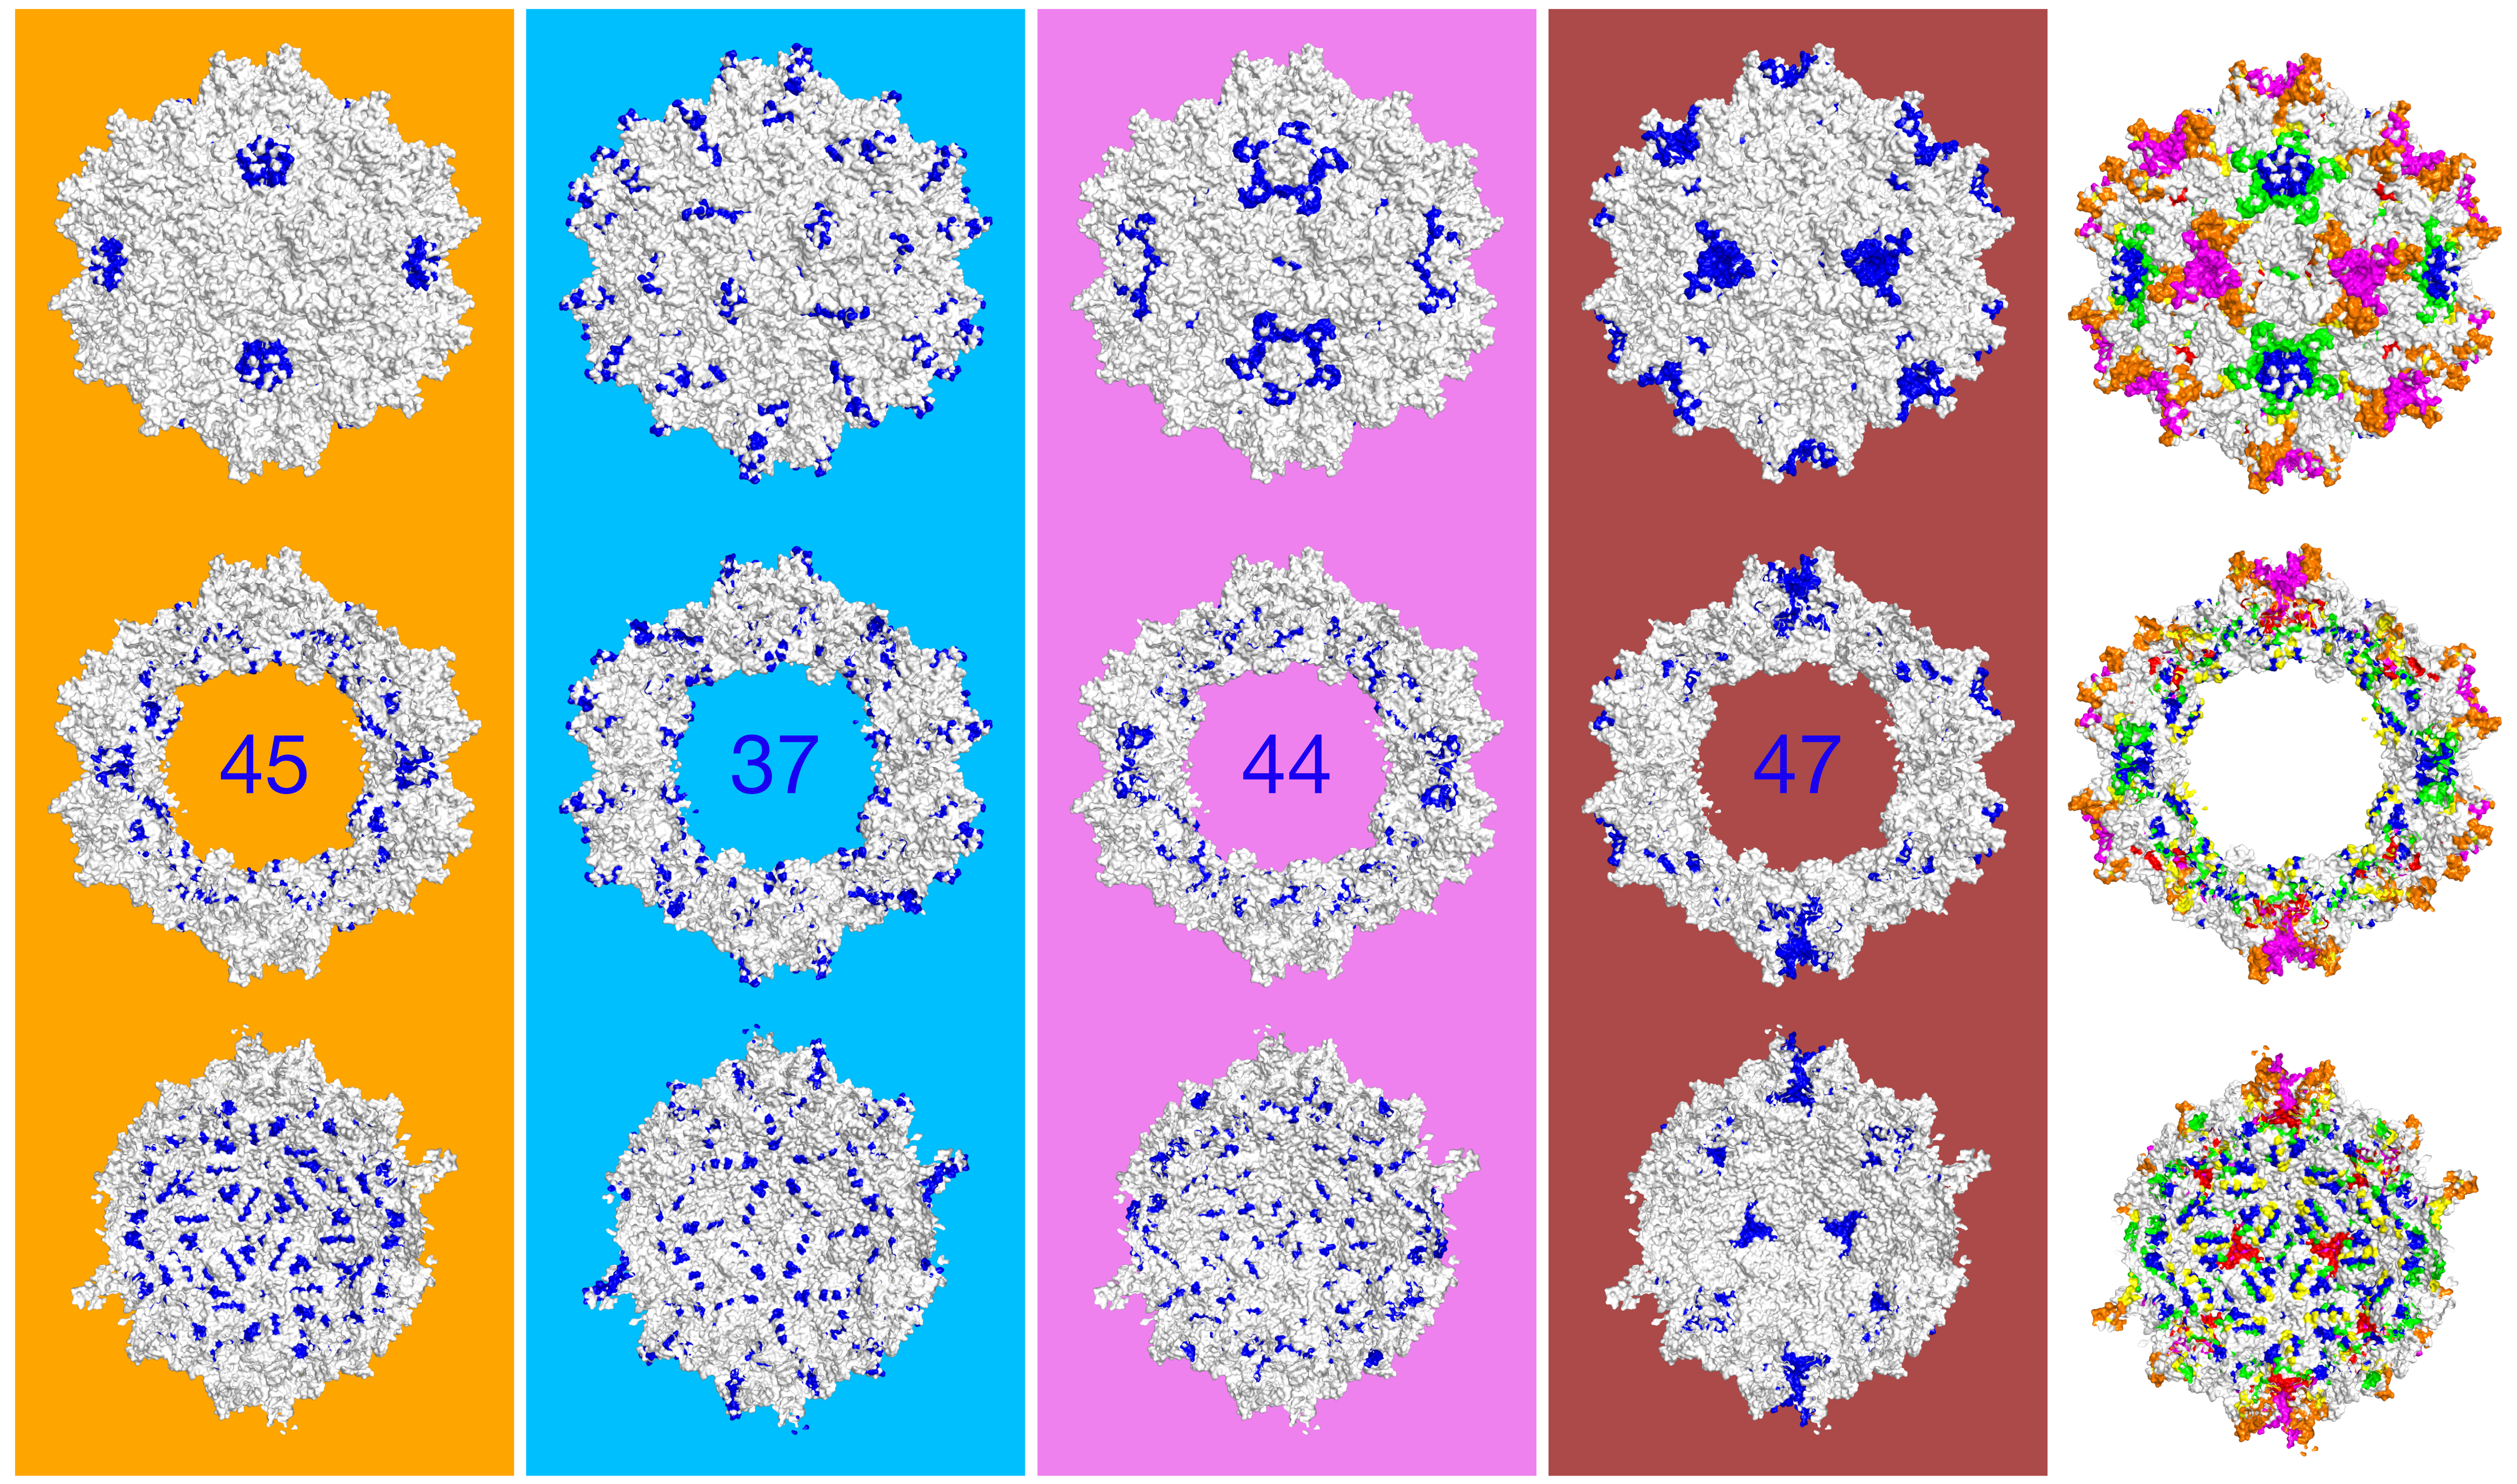
45
37
44
47

- Most generated variants are farther from other natural serotypes than AAV9 is (unlike reshuffling/recombination).
- While some regions are harder to diversify, we validate high edit distance mutations in all attempted regions.
- Distinct and structurally critical regions of the capsid are changed substantially (individual examples on the 1-4th right panels, aggregated mutations on the 5th panel).

Improved HEK293 transduction in Synthetic AAVs

- Many of the mutations were introduced in regions that are highly important for transduction.
- While generally speaking, higher edit distance does not improve transduction, many high edit distance variants transduce HEK293 cells better than AAV9.
- We show variants with more than 20 mutations introduced in the 570-620 region, exhibiting up to 96-fold improvement in transduction over AAV9.

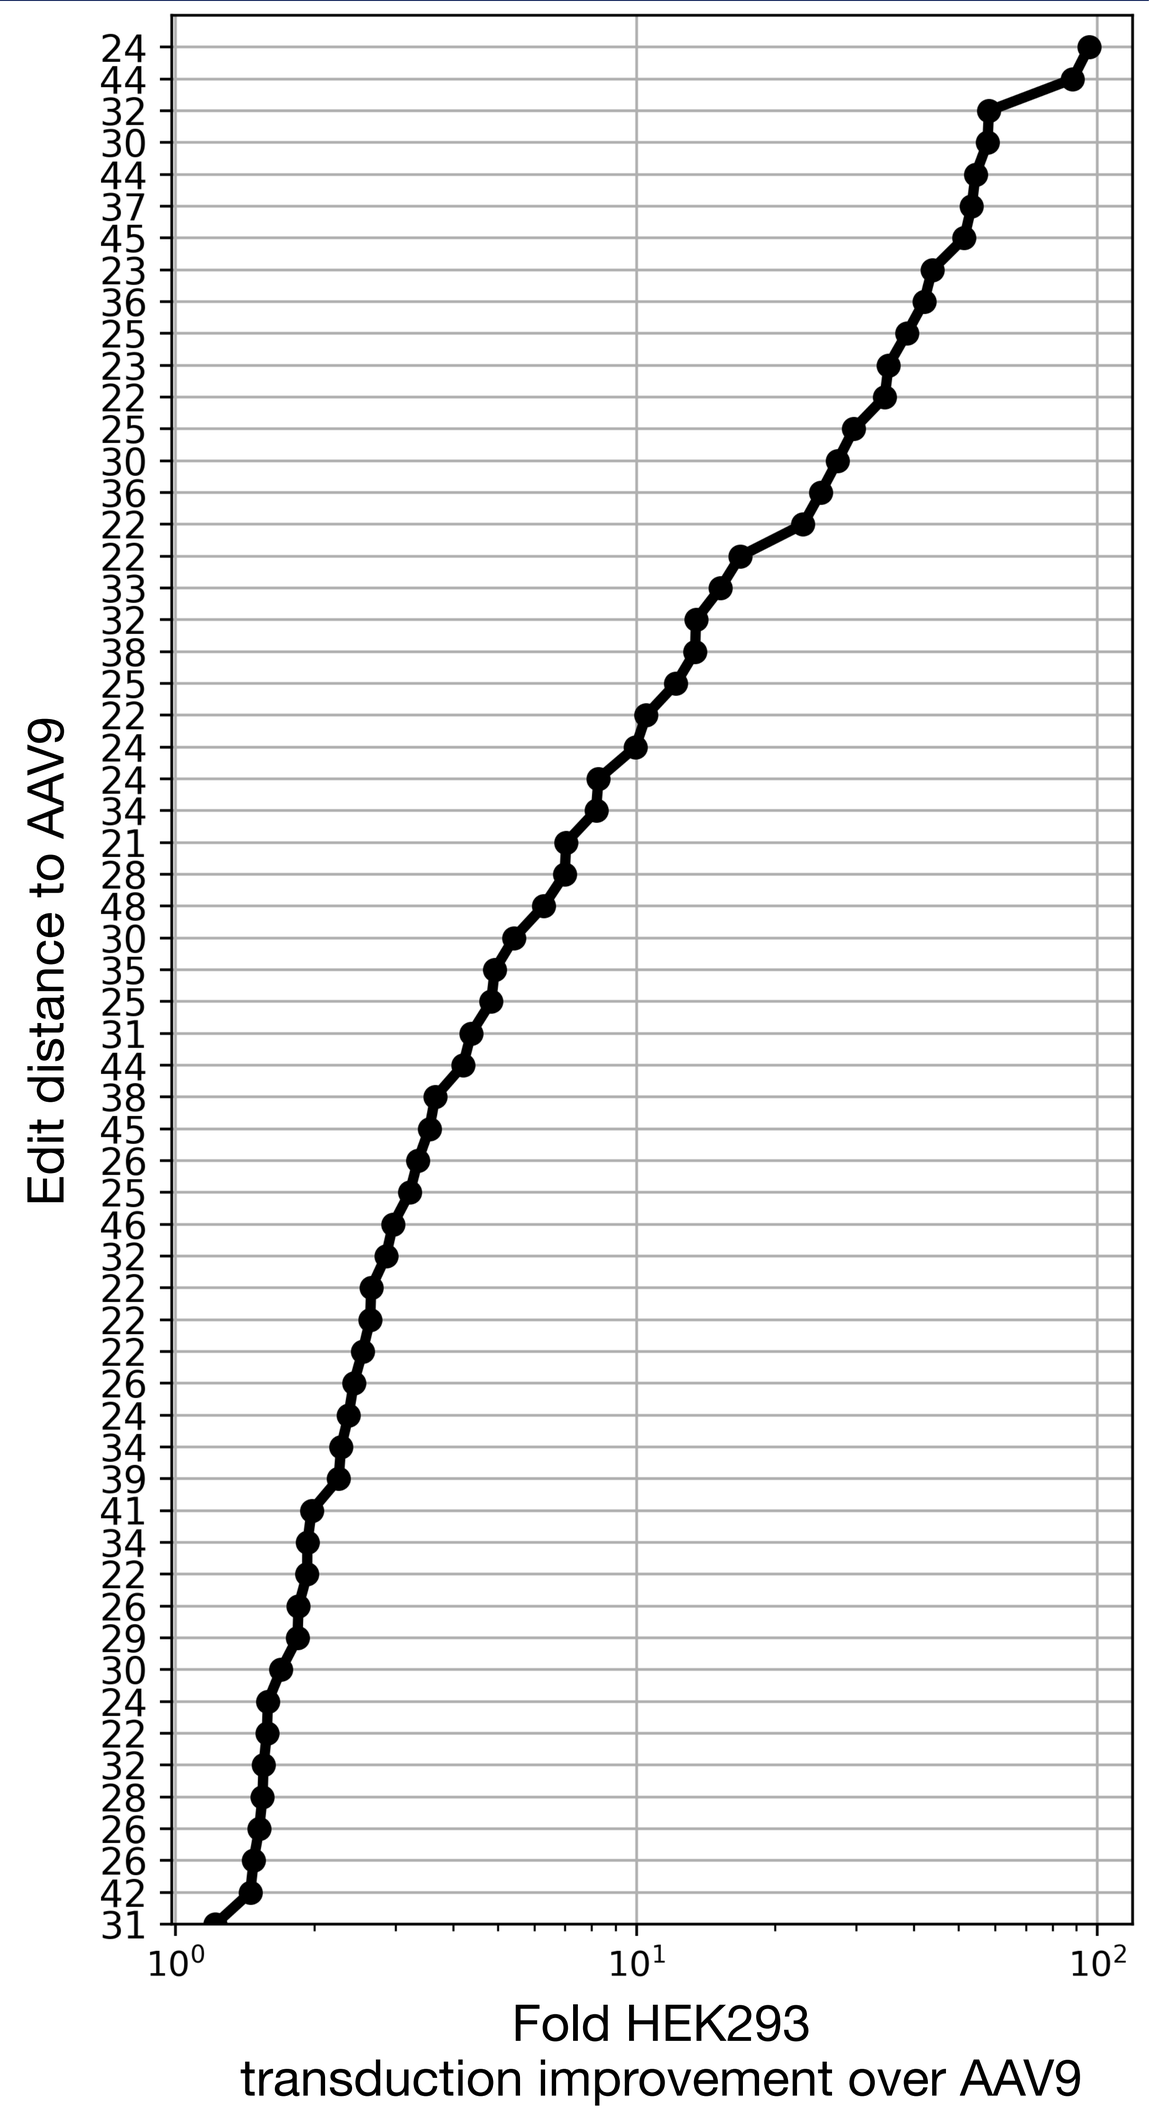
Edit distance to AAV9
Fold HEK293
transduction improvement over AAV9

Workflow

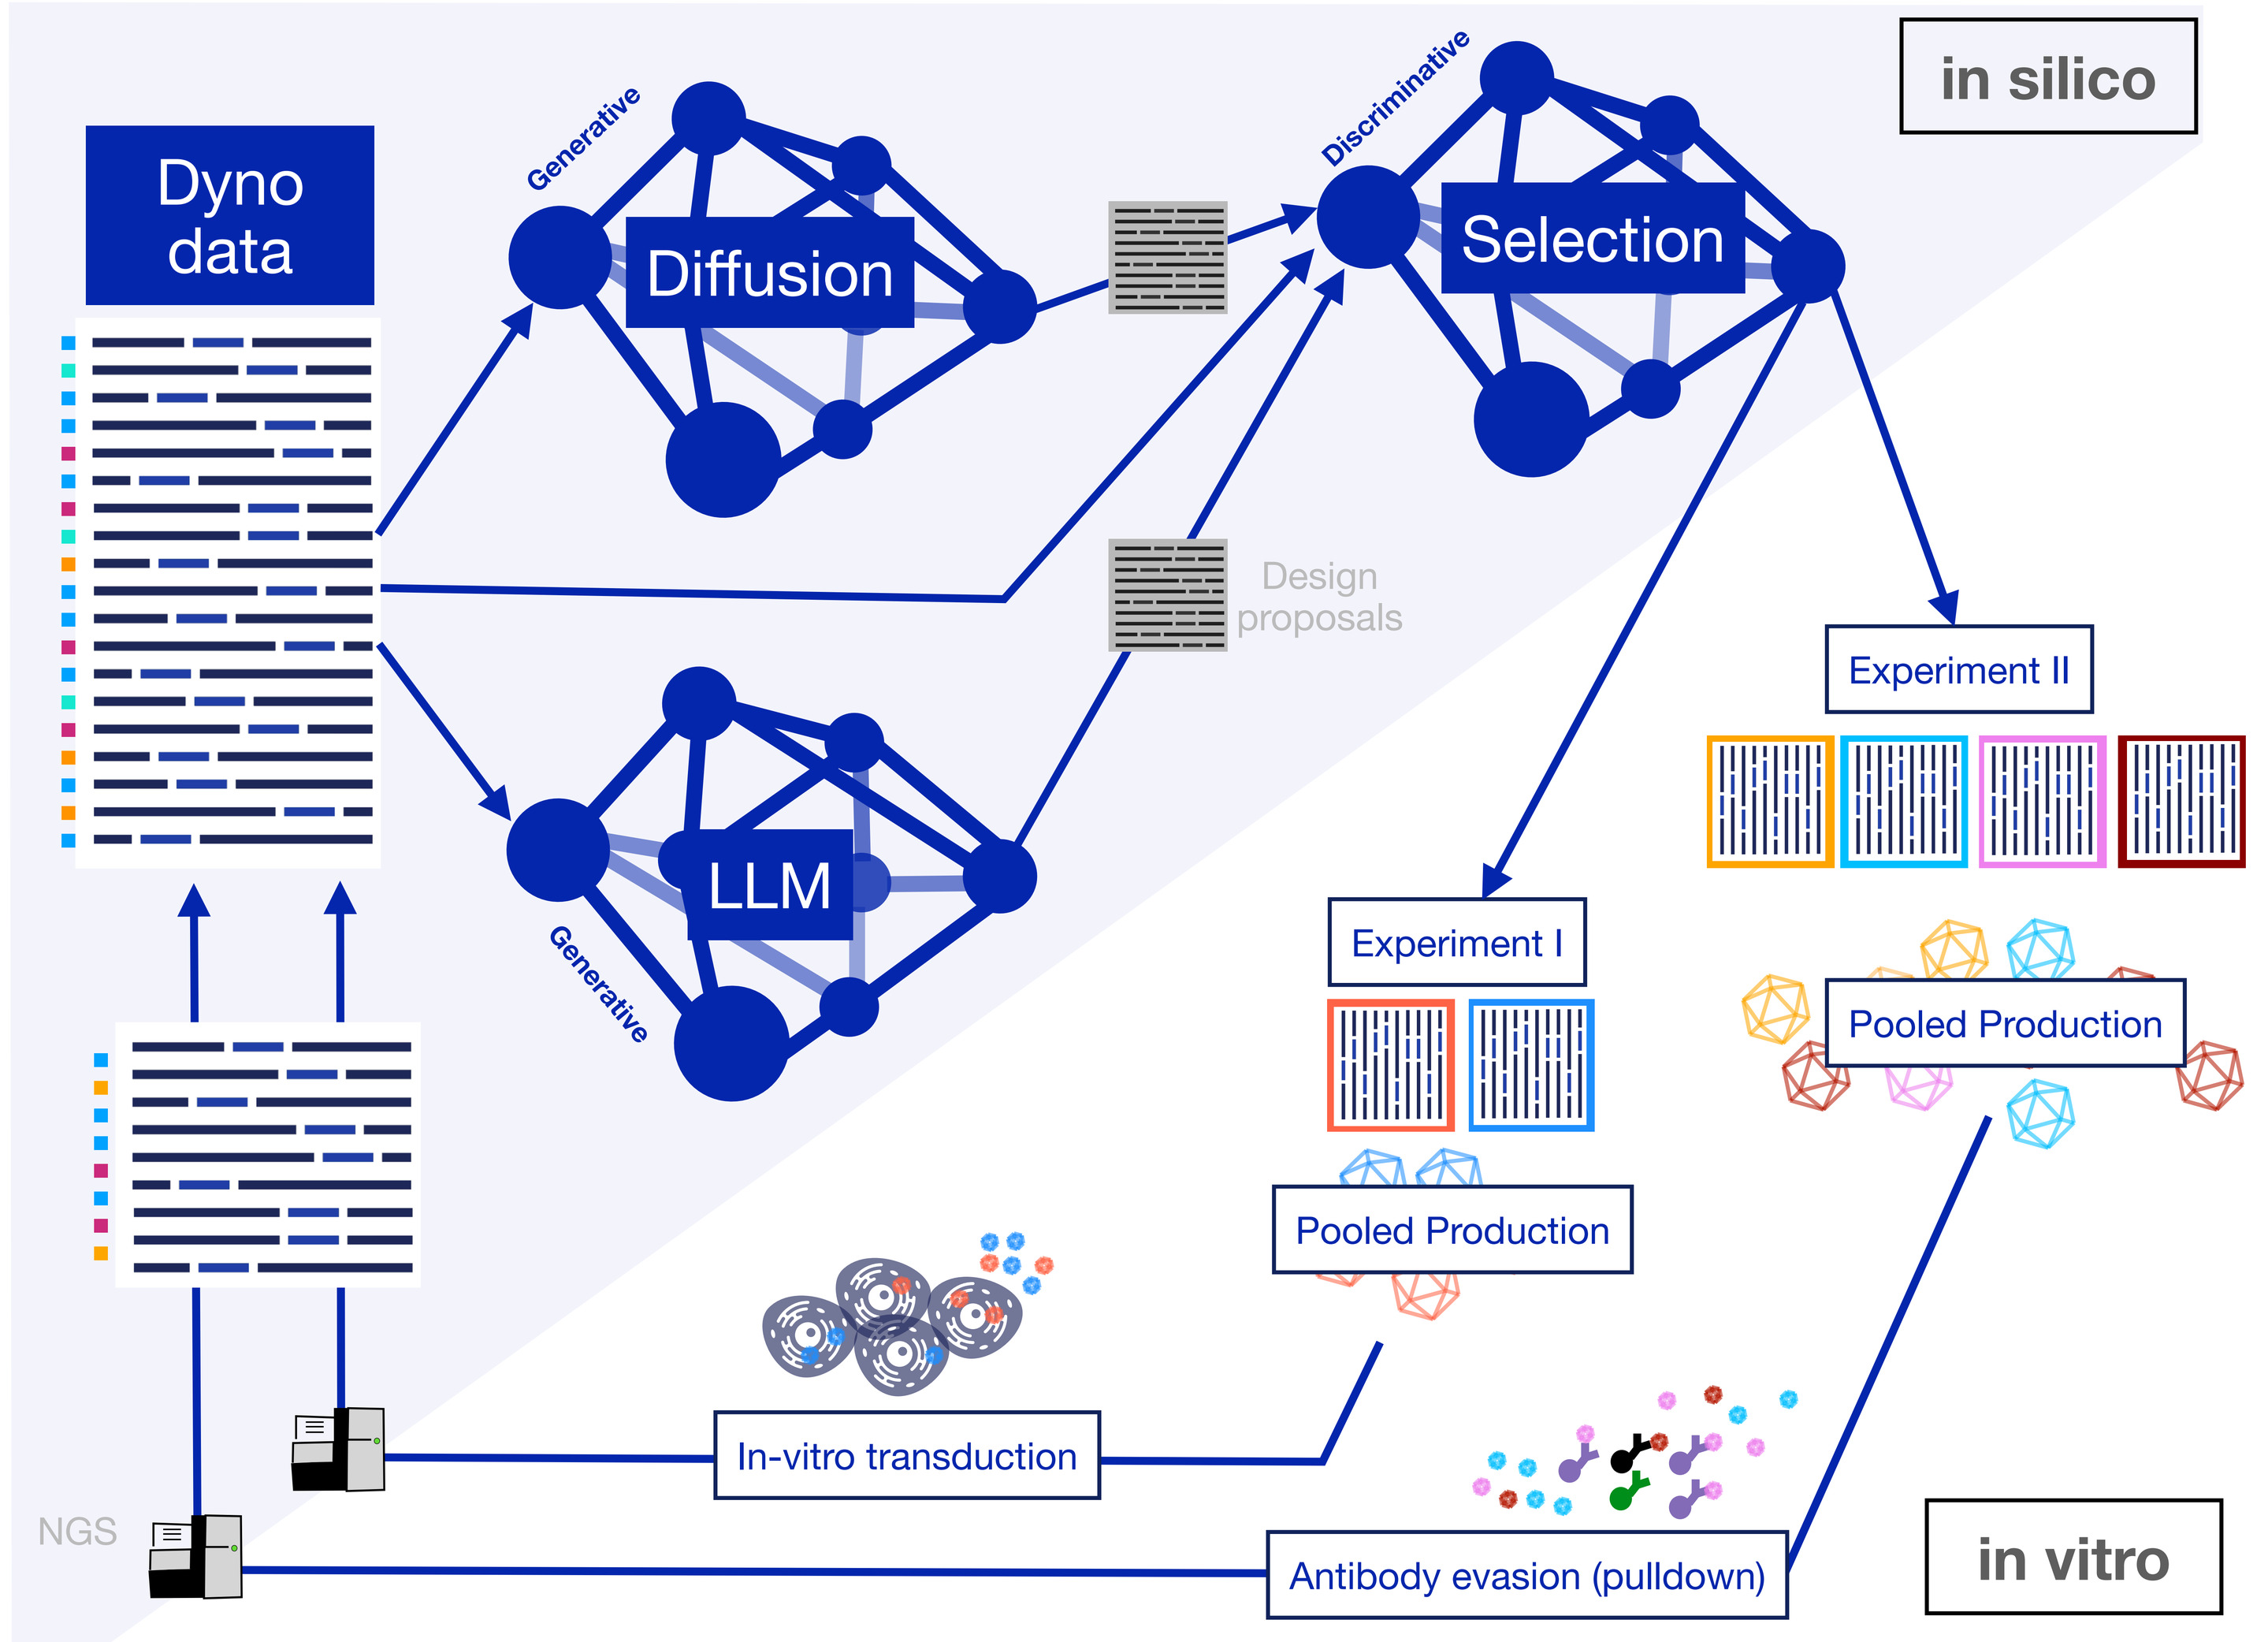
in silico
Dyno data
Generative
Diffusion
Discriminative
Selection
LLM
Generative
Design proposals
Experiment II
Experiment I
Pooled Production
Pooled Production
In-vitro transduction
Antibody evasion (pulldown)
NGS
in vitro

AI models successfully change conserved positions to unexpected residues

- The expected number of amino-acids seen in a position (perplexity) based on the *dependoparvovirus* Multiple Sequence Alignment (MSA).
- Mutations to residues not in the MSA results in higher surprise (Mutations to common residues are expected.)
- Single mutation effects per position shows conserved areas are hard to modify even by one mutation.

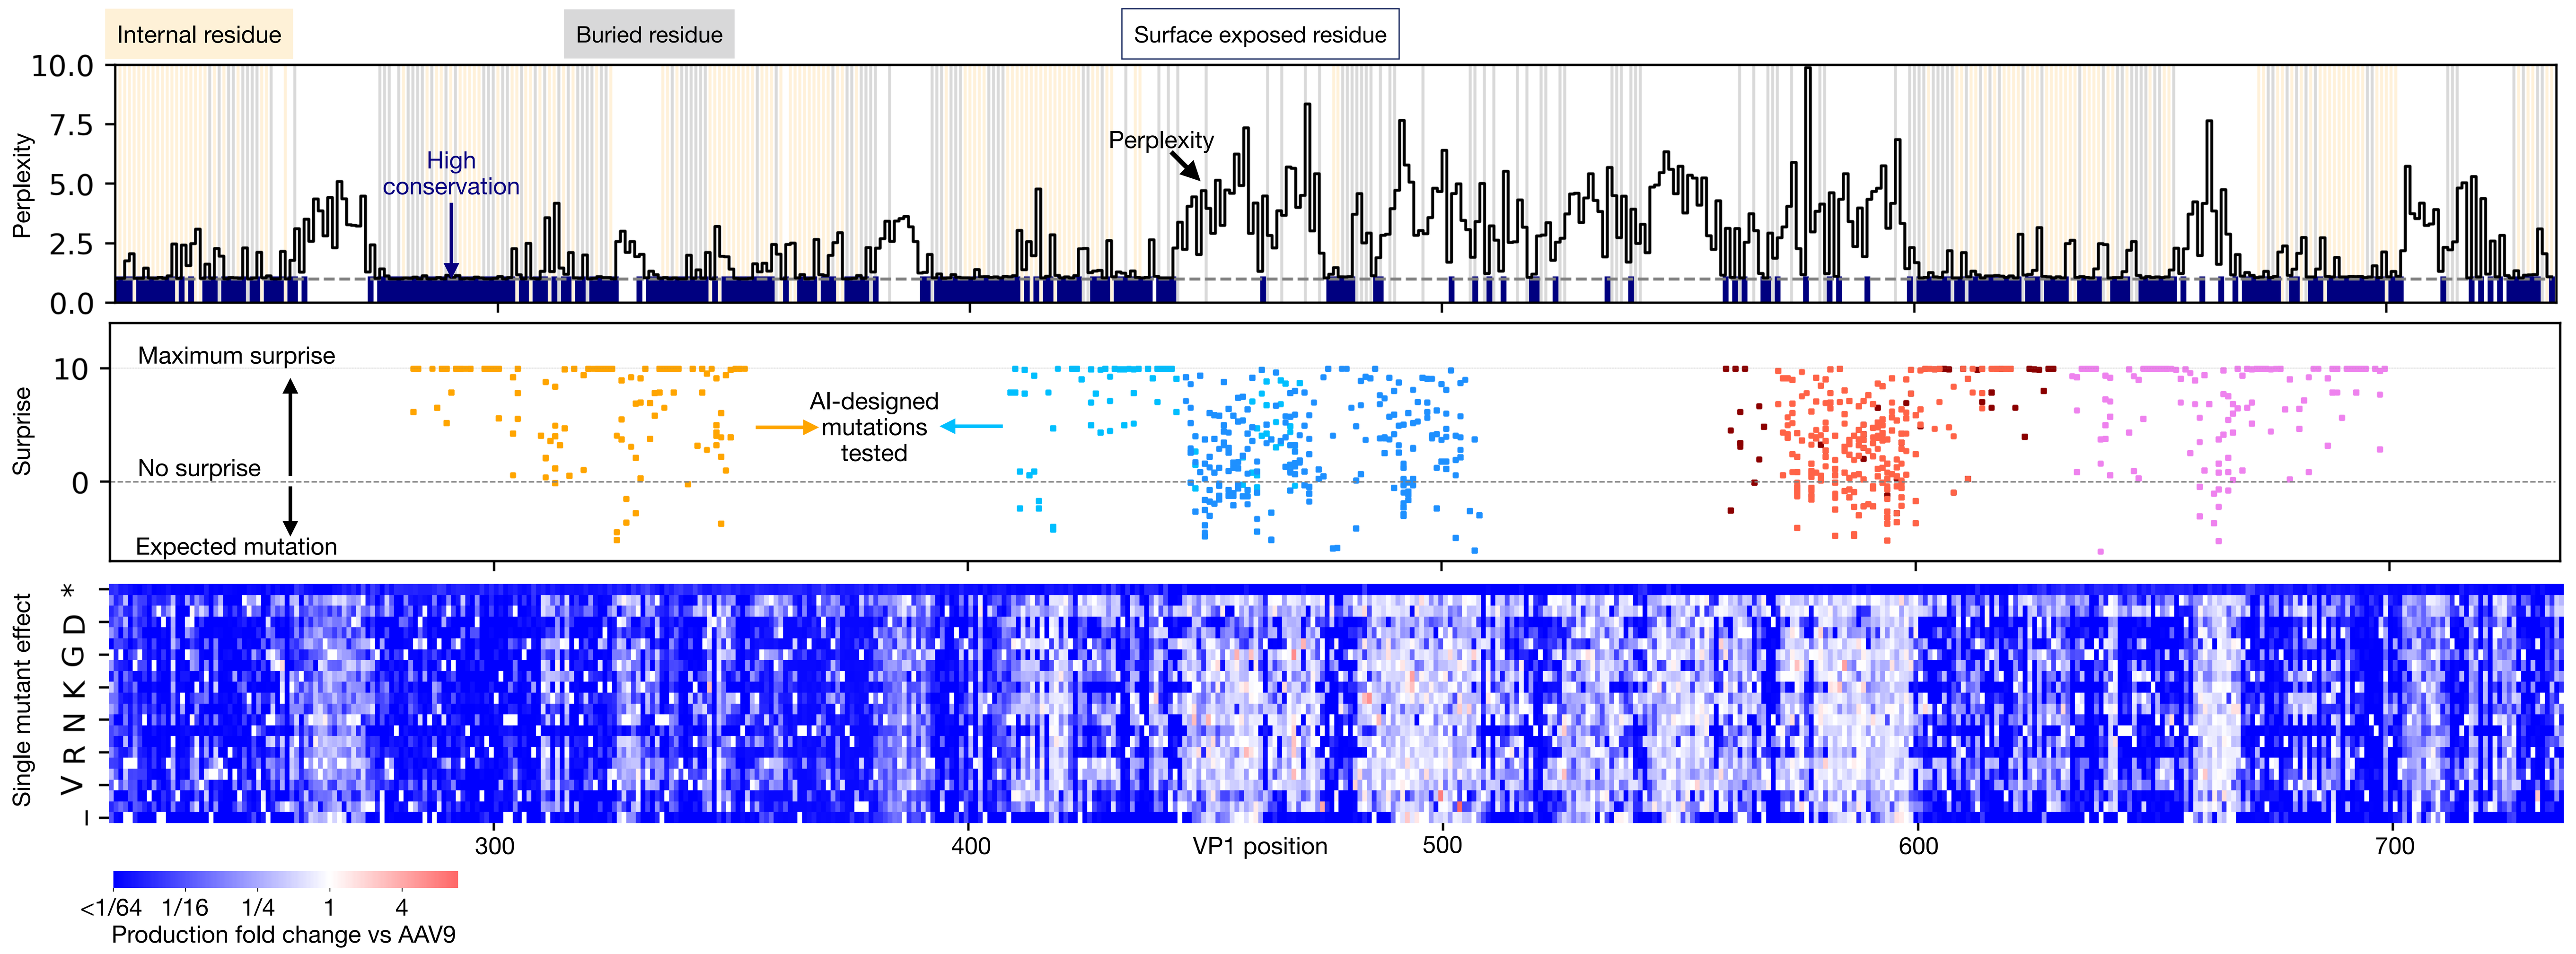
Internal residue
Buried residue
Surface exposed residue
Perplexity
High conservation
Maximum surprise
No surprise
Expected mutation
Surprise
AI-designed mutations tested
Single mutant effect
VP1 position
<1/64 1/16 1/4 1 4
Production fold change vs AAV9

Synthetic serotypes evade patient antibodies

- We find numerous variants with distinct structural modifications that evade recognition by serum antibodies (tested at a 1:10 dilution) in 12 individuals with pre-existing anti-AAV9 neutralizing antibody titers.
- These variants also evade recognition by pooled patient IVIG at 1:20 dilution and 4 monoclonal antibodies targeting distinct structural features of the capsid.

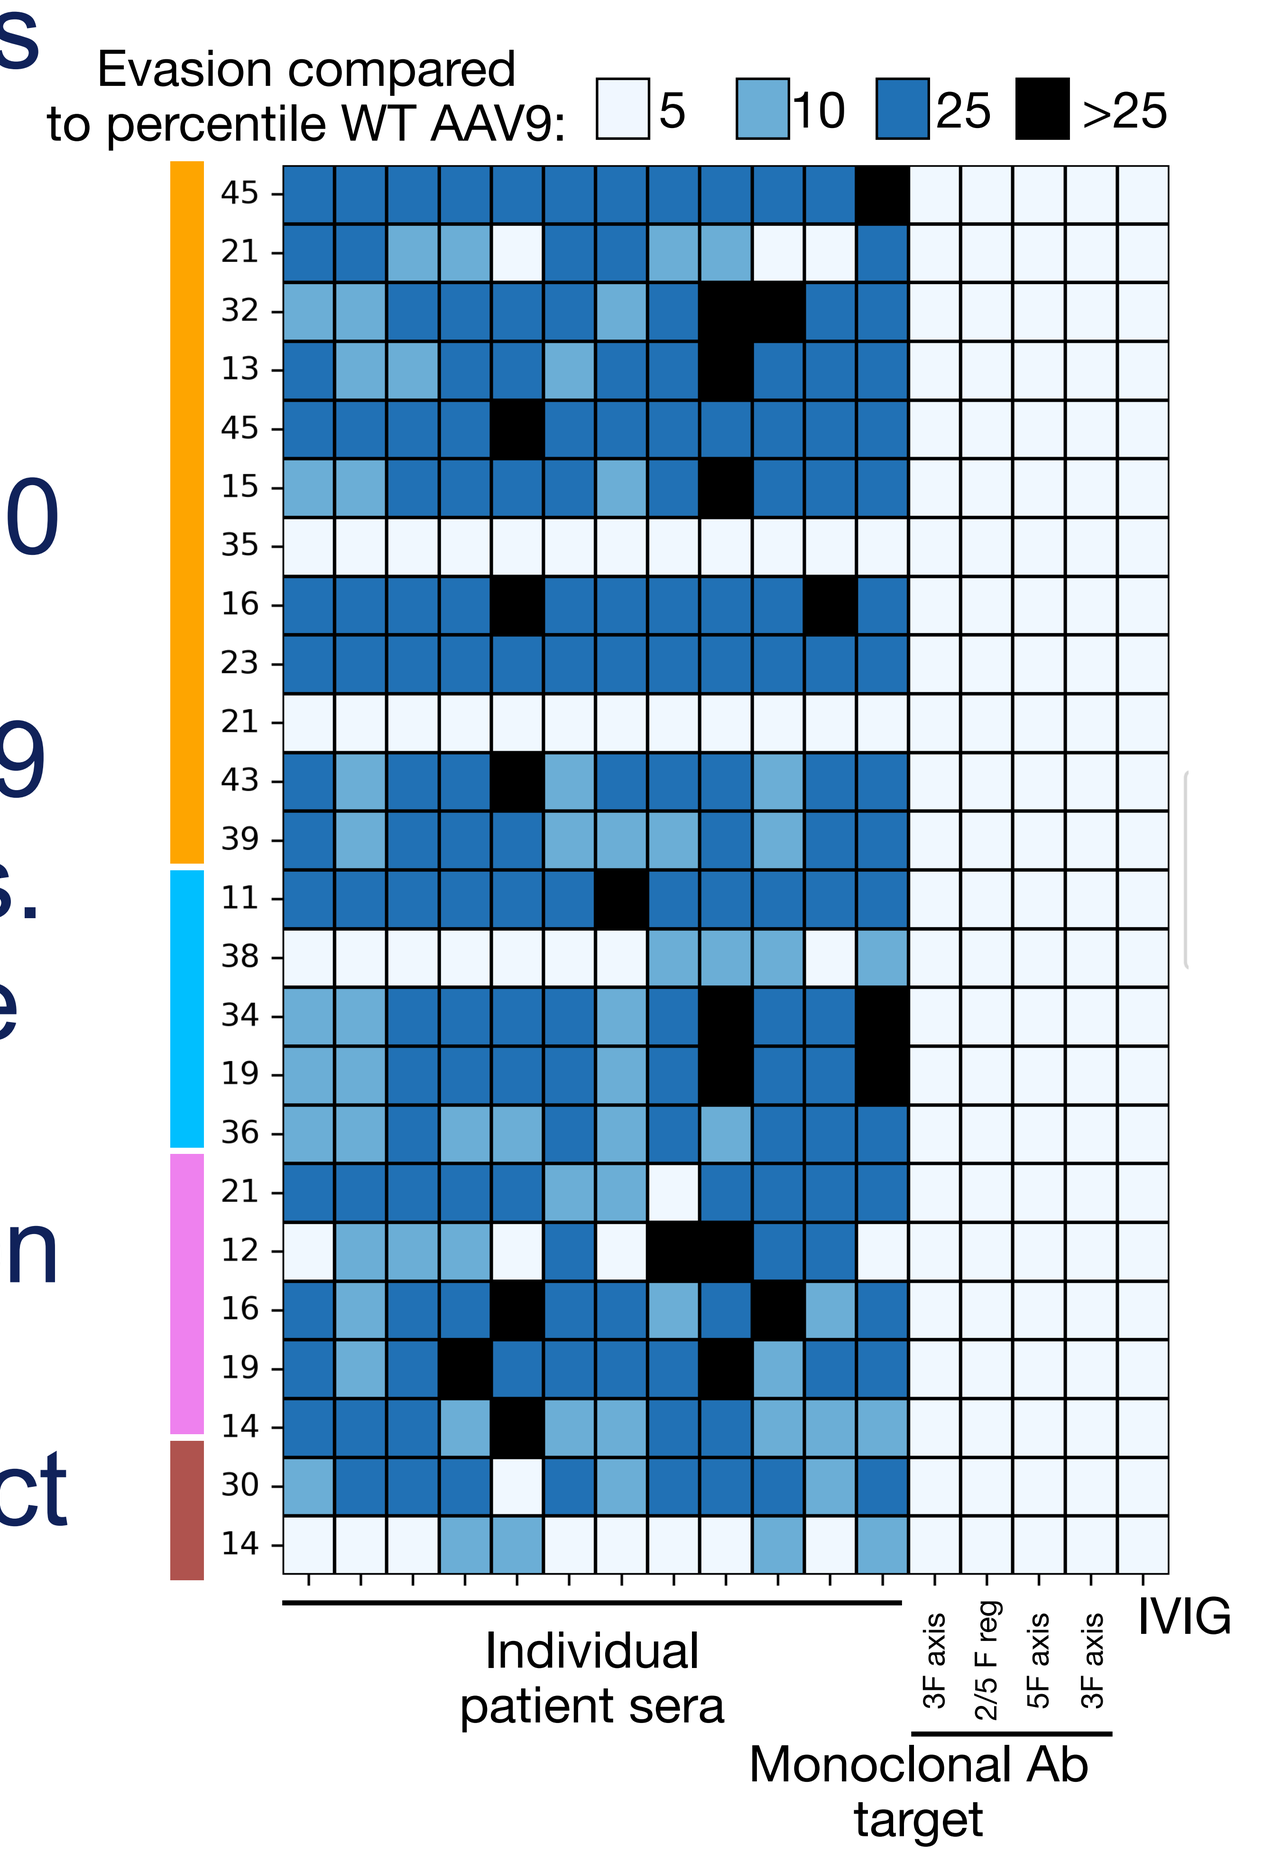
Evasion compared to percentile WT AAV9:
5
10
25
>25
Individual patient sera
3F axis
2/5 F reg
5F axis
3F axis
Monoclonal Ab target
IVIG